Evaluation of Tumor Microenvironment Identifies Immune Correlates of Response to Combination Immunotherapy with Margetuximab (M) and Pembrolizumab (P) in HER2+ Gastroesophageal Adenocarcinoma (GEA)

JOHN VAN GEEST CANCER RESEARCH CENTRE
Nottingham Trent University

Abstract #2547

S. Rutella[1], S.E. Church[2], J. Vadakekolathu[1], S. Reeder[1], A. Sullivan[2], S. Warren[2], J. Baughman[3], J. Muth[3], H. Park[4], H. Uronis[5], Y-K. Kang[6], M. Ng[7], P. Enzinger[8], K.W. Lee[9], K. Huber[3], A. Wynter-Horton[3], D. Li[3], Y-J. Bang[10], J. Davidson-Moncada[3], D. Catenacci[11]

[1]John van Geest Cancer Research Centre, Nottingham Trent University, United Kingdom; [2]NanoString Technologies Inc., Seattle, WA; [3]MacroGenics Inc., Rockville, MD; [4]Washington University School of Medicine, St. Louis, MO; [5]Duke University Medical Center, Durham, NC; [6]Asan Medical Center, Seoul, South Korea; [7]National Cancer Centre Singapore, Singapore; [8]Dana-Farber Cancer Institute, Boston, MA; [9]Seoul National University Bundang Hospital, Seongnam, South Korea; [10]Seoul National University Hospital, Seoul, South Korea; [11]The University of Chicago Medical Center and Biological Sciences, Chicago, IL

NCT02689284

Background

- Gastric cancer is the fifth most common cancer and the third most common cause of cancer deaths worldwide
- Despite improvements in treatment, the 5-year survival of patients with GEA is disappointing
- Individual molecular subtypes of GEA display preferential responses to PD-1 blockade
- Margetuximab is an investigational Fc-optimized anti-HER2 monoclonal antibody being tested in combination with pembrolizumab in HER2+ GEA post trastuzumab

Materials and Methods

- 92 patients with advanced, relapsed/refractory HER2+ gastric cancer (GC) and gastroesophageal junction (GEJ) cancer were treated with margetuximab + pembrolizumab in the CP-MGAH22-05 study
- 55 pre-treatment tumor samples were assessed by NanoString's PanCancer IO360™ (IO360) gene expression assay (for research use only)[1]
 - IO360 signature scores were calculated and are presented as fold changes (FC) and analyzed by unpaired t test
- Associations examined include:
 - Overall inflamed tumor microenvironment (TME)
 - Complete response (CR) n=1, partial response (PR) n=13, stable disease (SD) n=23, progressive disease (PD) n=18
 - Immunohistochemistry (IHC) status for PD-L1 (positive n=25, versus negative n=27) and HER2 (IHC3+ n=45, versus IHC2+ n=10)
 - Tumor location (GC n=46, versus GEJ cancer, n=9)

Results

Best Response by HER2 Expression and Tumor Site

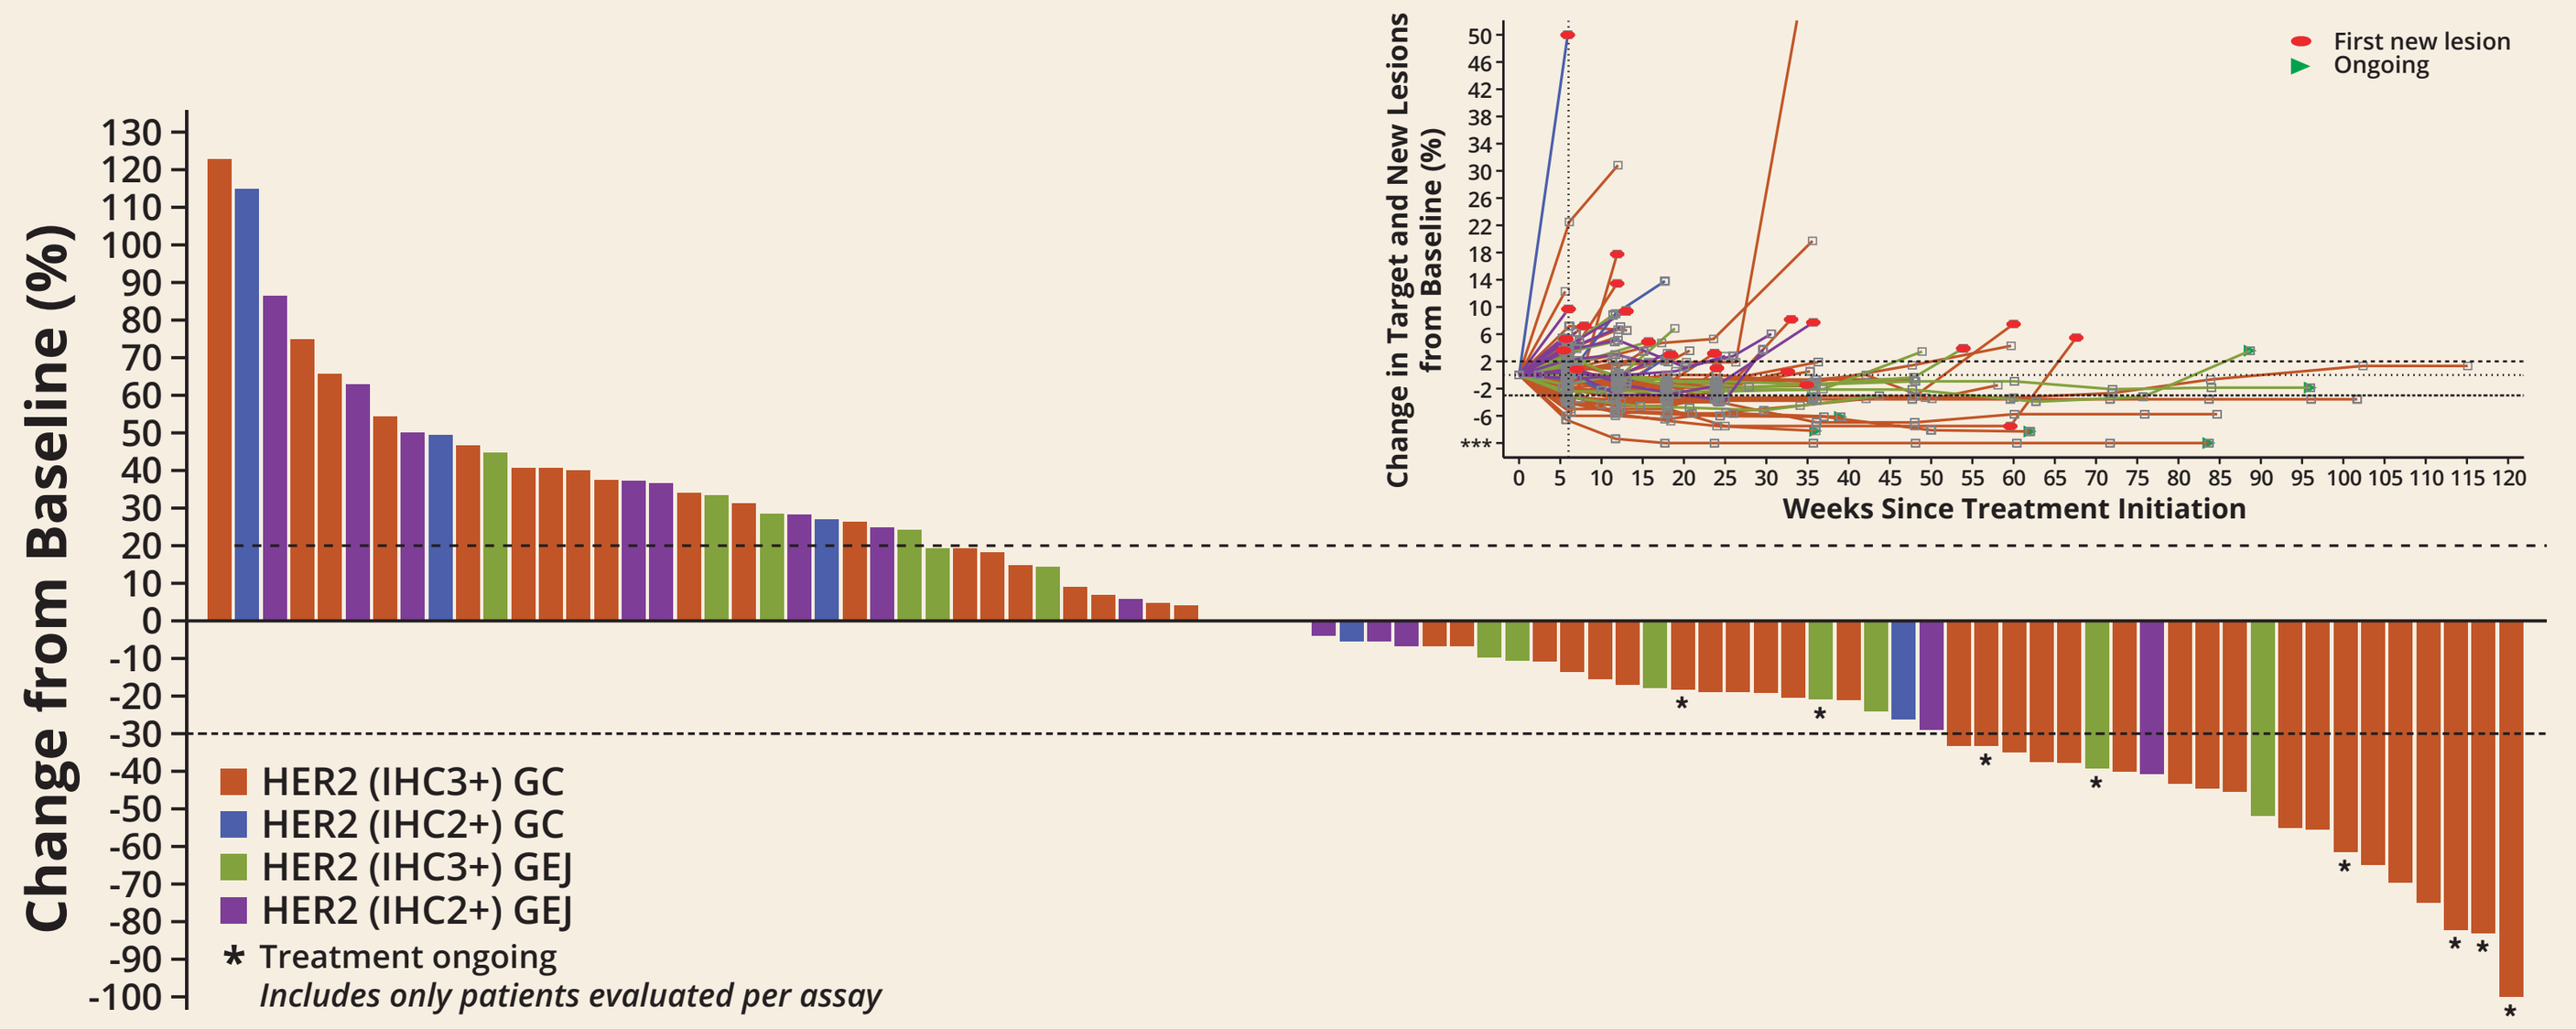
Change from Baseline (%)
HER2 (IHC3+) GC
HER2 (IHC2+) GC
HER2 (IHC3+) GEJ
HER2 (IHC2+) GEJ
* Treatment ongoing
Includes only patients evaluated per assay
Change in Target and New Lesions from Baseline (%)
Weeks Since Treatment Initiation
First new lesion
Ongoing

HER2 (IHC3+) Gastric Cancer

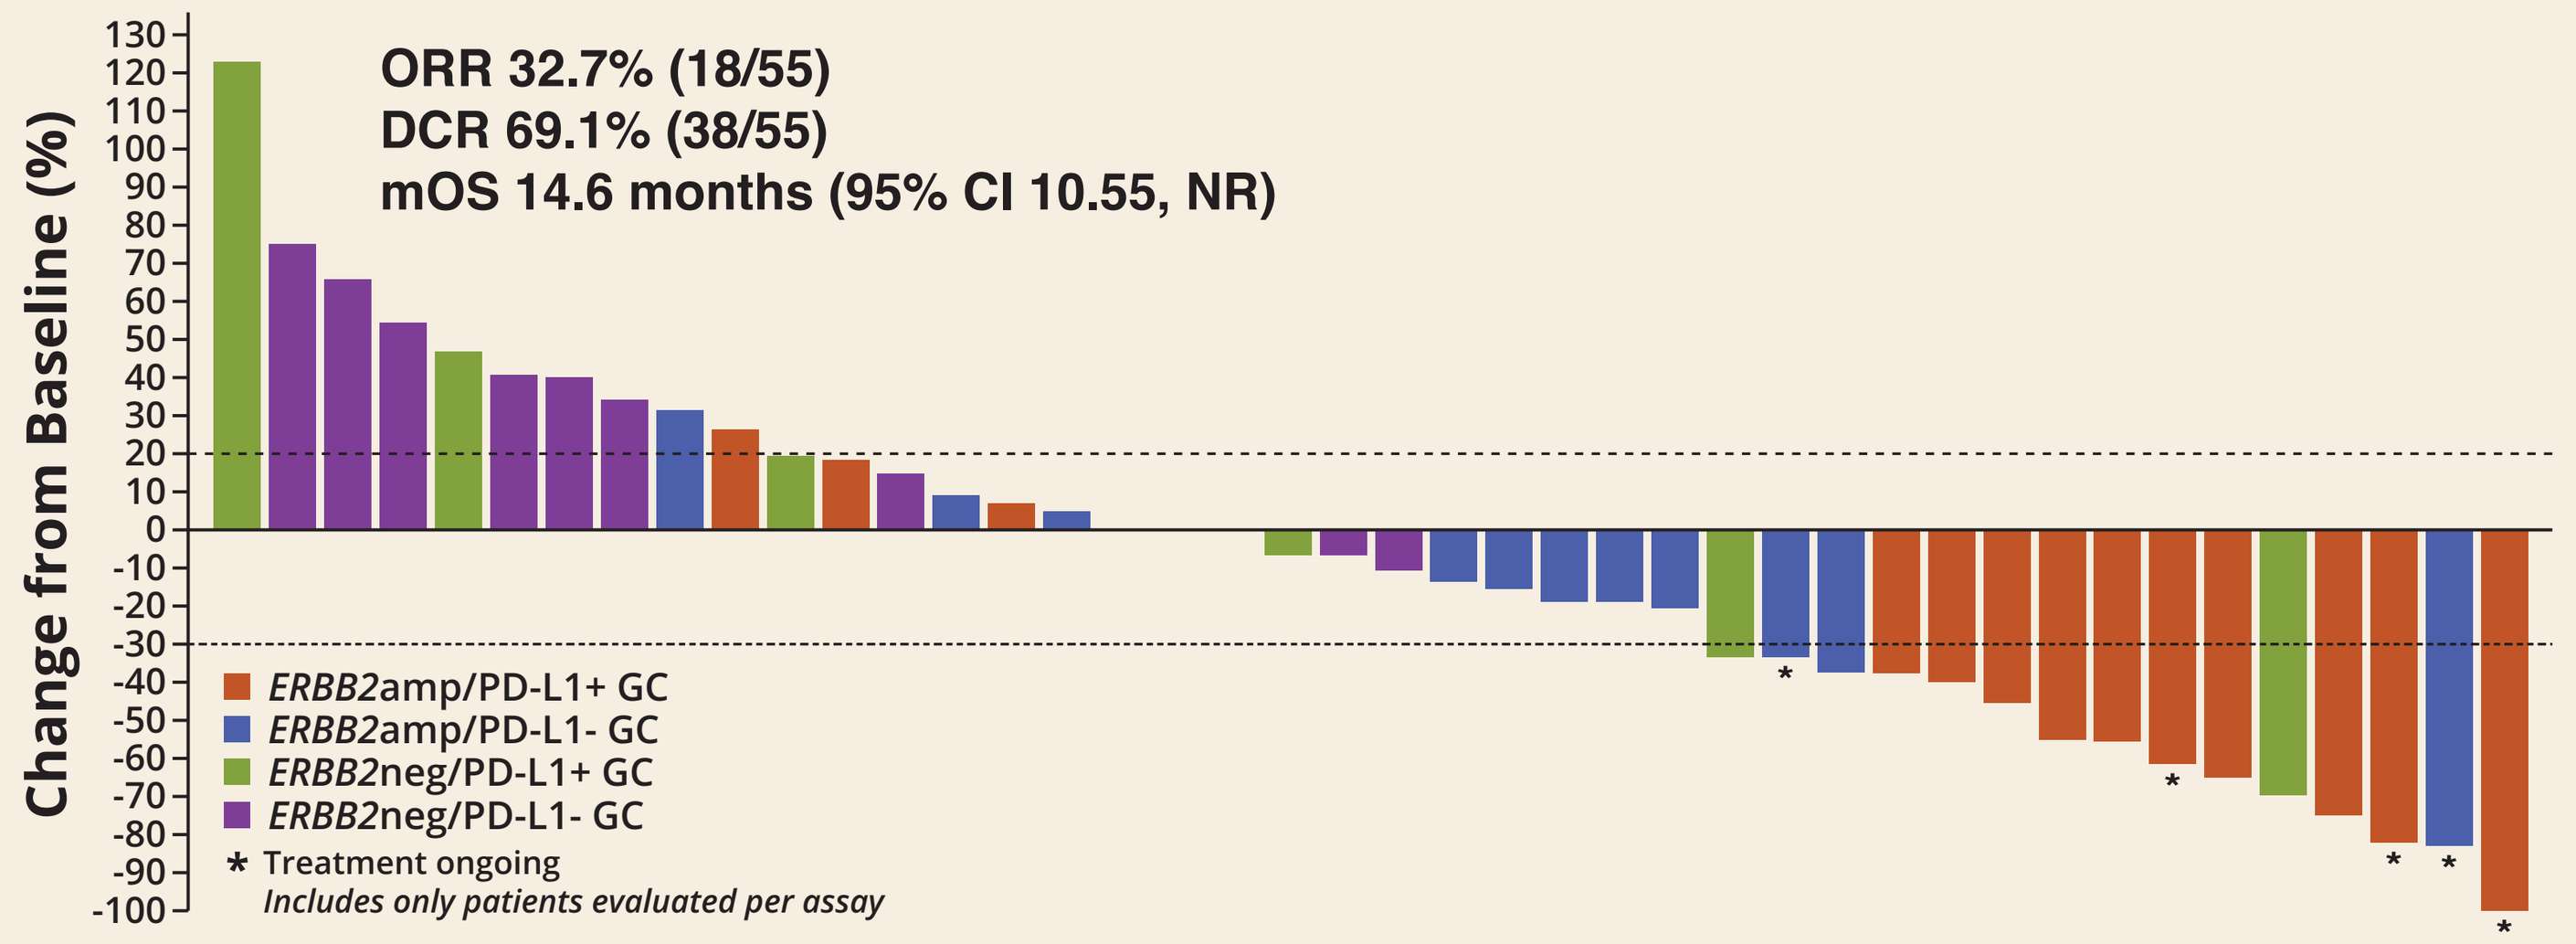
ORR 32.7% (18/55)
DCR 69.1% (38/55)
mOS 14.6 months (95% CI 10.55, NR)
Change from Baseline (%)
ERBB2amp/PD-L1+ GC
ERBB2amp/PD-L1- GC
ERBB2neg/PD-L1+ GC
ERBB2neg/PD-L1- GC
* Treatment ongoing
Includes only patients evaluated per assay

- Increased response rate in patients with *ERBB2*amp, a surrogate for HER2 expression, post trastuzumab
- Double positive population (*ERBB2*amp/PD-L1+) most sensitive population to combination therapy

ERBB2/HER2 Expression is Associated with Response

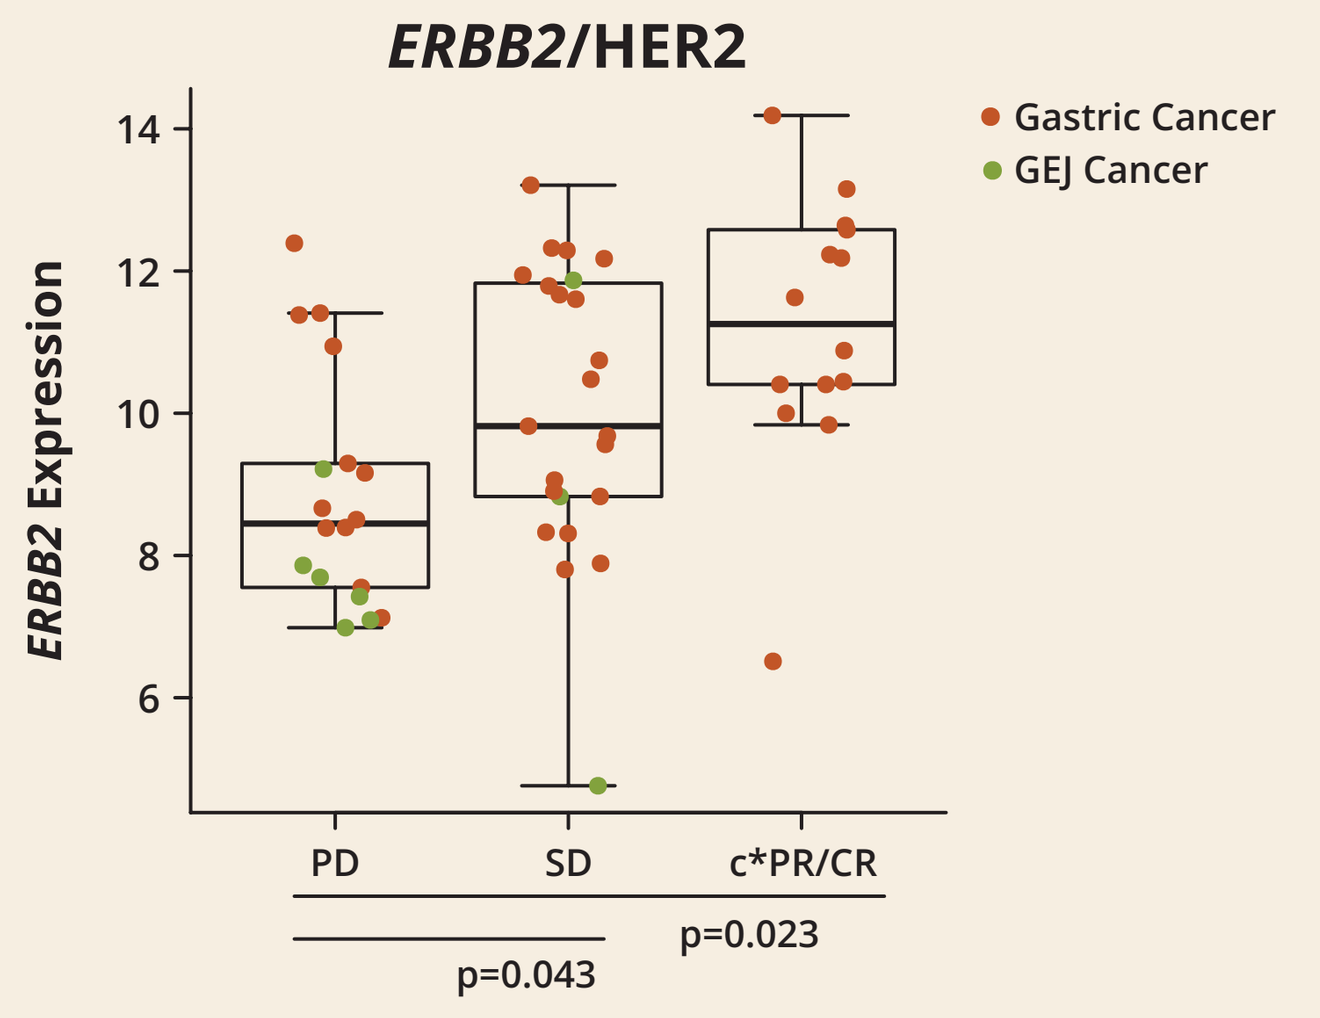
ERBB2/HER2
ERBB2 Expression
Gastric Cancer
GEJ Cancer
PD
SD
c*PR/CR
p=0.043
p=0.023

- *ERBB2* mRNA expression significantly correlated with response, being 5.13 times higher in patients achieving CR/PR compared with patients with PD

*c: Confirmed

Differential Expression Associated with Anti-Tumor Activity

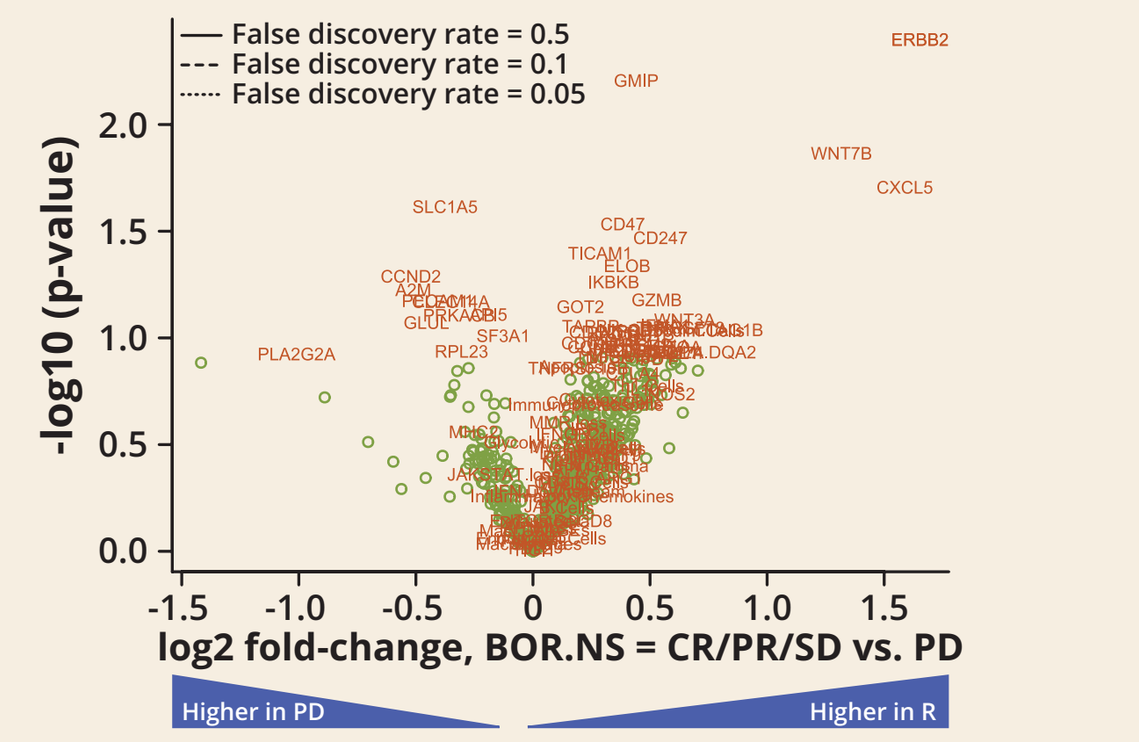
False discovery rate = 0.5
False discovery rate = 0.1
False discovery rate = 0.05
-log10 (p-value)
log2 fold-change, BOR.NS = CR/PR/SD vs. PD
Higher in PD
Higher in R

Gene	Fold Change	SE	p-value
ERBB2	3.145	0.549	0.004
GMIP	1.357	0.155	0.006
WNT7B	2.493	0.516	0.014
CXCL5	3.006	0.660	0.020
SLC1A5	0.771	0.162	0.024
CD47	1.305	0.171	0.029
CD247	1.459	0.250	0.034
TICAM1	1.220	0.136	0.040
ELOB	1.321	0.197	0.046
CCND2	0.697	0.262	0.051

- *ERBB2* mRNA expression significantly correlated with anti-tumor activity, being 3.1 times higher (p=0.004) in patients achieving CR/PR/SD compared with patients with PD

ERBB2/HER2 Expression is Associated with Anti-Tumor Activity

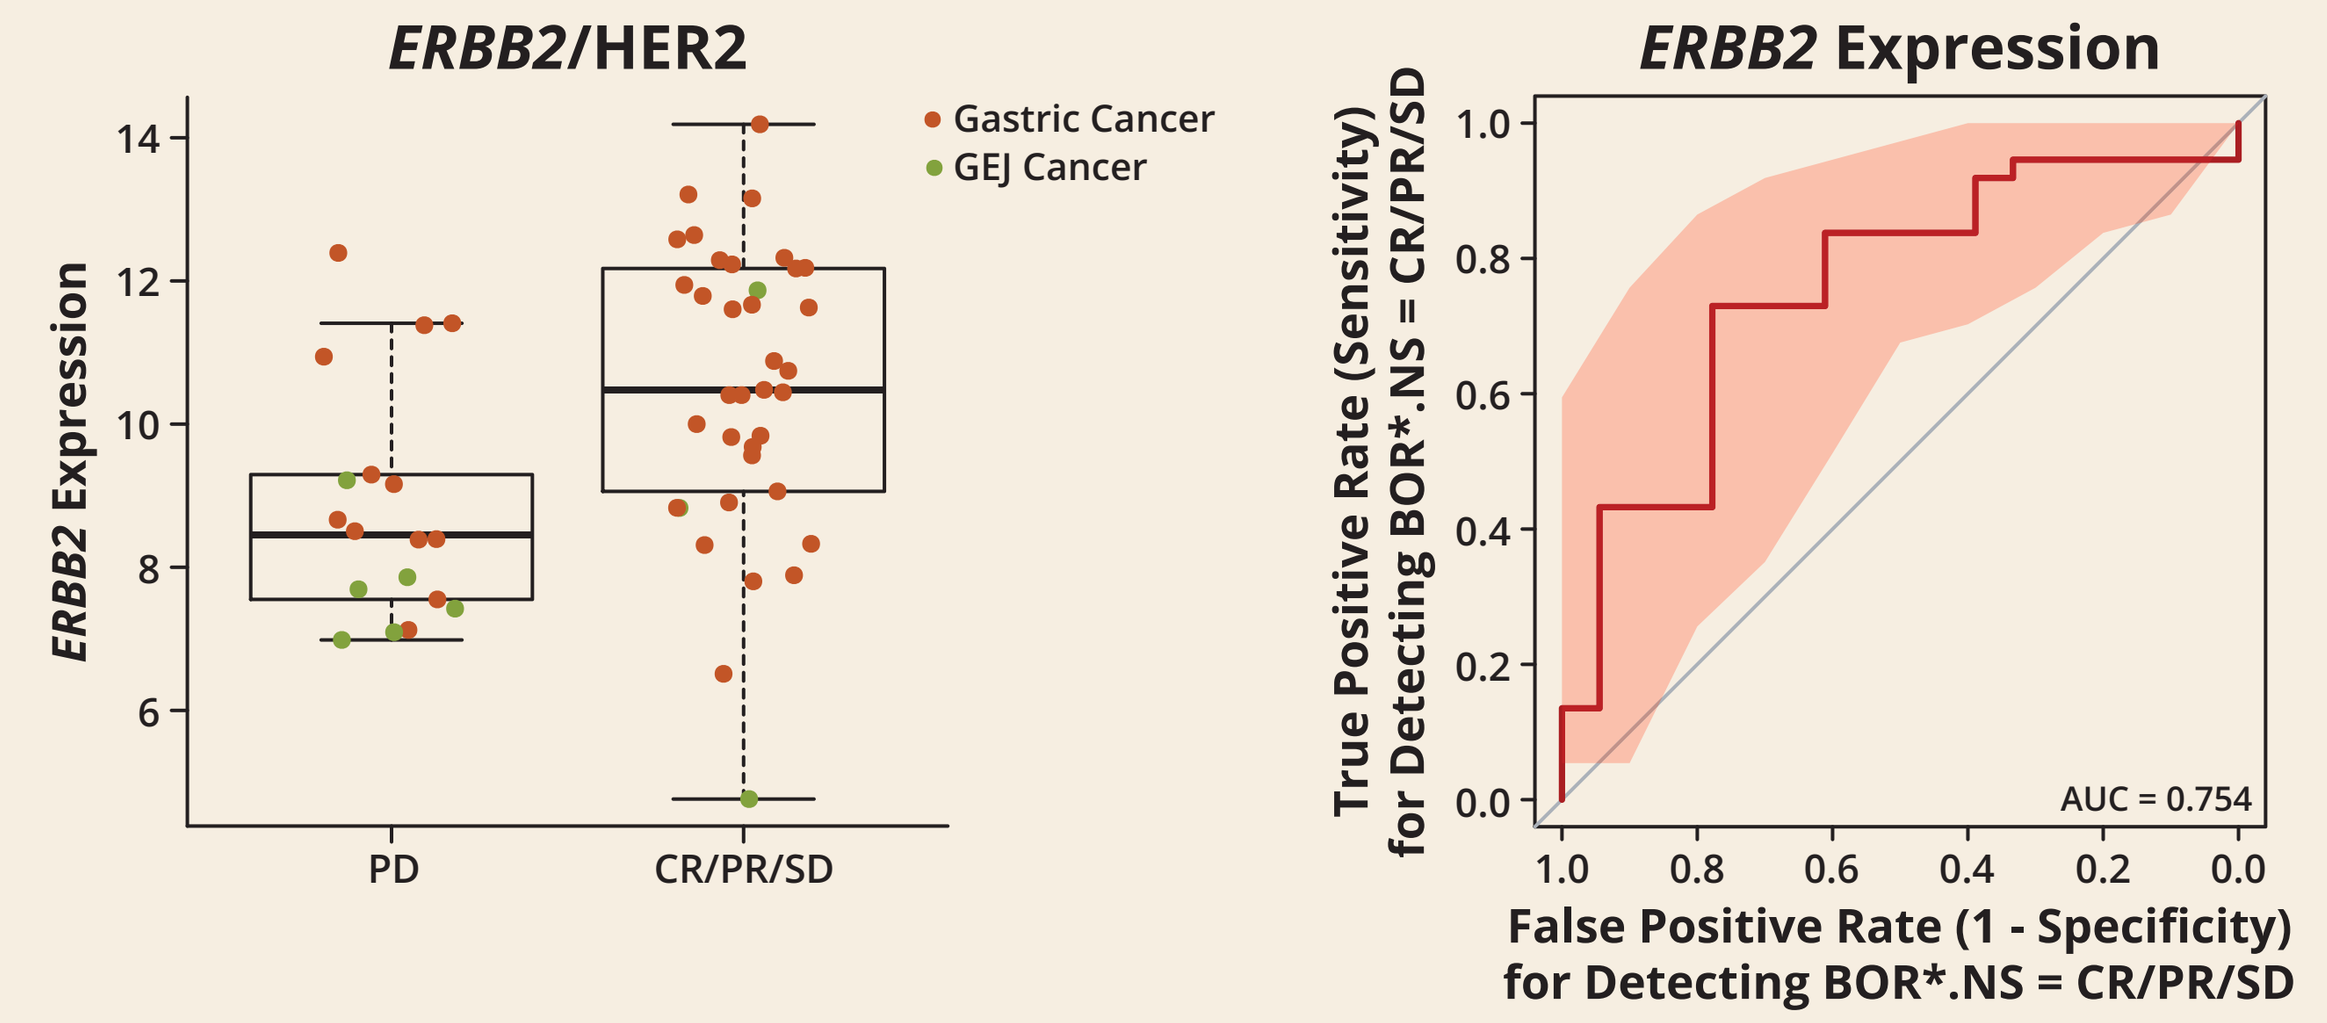
ERBB2/HER2
ERBB2 Expression
Gastric Cancer
GEJ Cancer
PD
CR/PR/SD
ERBB2 Expression
True Positive Rate (Sensitivity) for Detecting BOR*.NS = CR/PR/SD
False Positive Rate (1 - Specificity) for Detecting BOR*.NS = CR/PR/SD
AUC = 0.754

- *ERBB2* expression was associated with anti-tumor activity of margetuximab + pembrolizumab with an AUC of 0.754

*Best overall response

NK CD56dim Cell Abundance Trends Higher with Anti-Tumor Activity

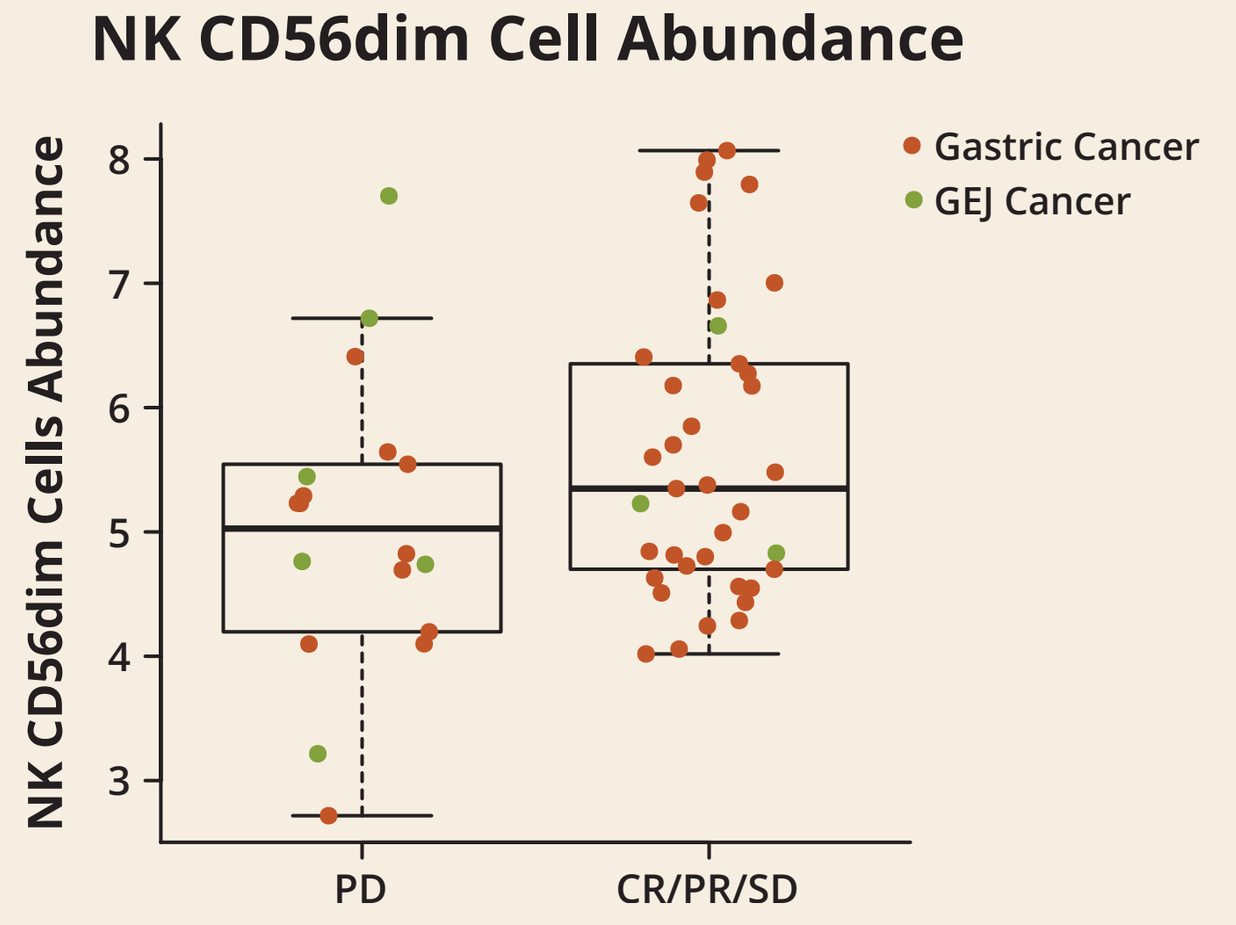
NK CD56dim Cell Abundance
NK CD56dim Cells Abundance
Gastric Cancer
GEJ Cancer
PD
CR/PR/SD

NK CD56dim Genes
IL21R
KIR2DL3
KIR3DL1
KIR3DL2

- Tumor control was associated with a modest increase in CD56dim NK cells (p=0.092)

Baseline HER2 Immunohistochemistry

HER2 IHC PFS	HER2 IHC2+ (n=21)	HER2 IHC3+ (n=71)
# of events	21	57
Median PFS (95% CI)	1.38 months (1.31–1.61)	4.34 months (2.60–5.62)
HR by Cox model, 0.32; 95% CI, 0.17–0.53; Log-rank P<0.0001		
6-month PFS rate (95% CI)	4.8% (0.3–19.7)	38% (26.4–49.5)
12-month PFS rate (95% CI)	NA	24% (13.8–35.0)

HER2 IHC OS	HER2 IHC2+ (n=21)	HER2 IHC3+ (n=71)
# of events	19	37
Median OS (95% CI)	5.6 months (2.99–11.53)	13.9 months (10.55–20.47)
HR by Cox model, 0.30; 95% CI, 0.17–0.53; Log-rank P<0.0001		
12-month OS rate (95% CI)	24.4% (8.2–45.3)	59.8% (44.6–70.8)

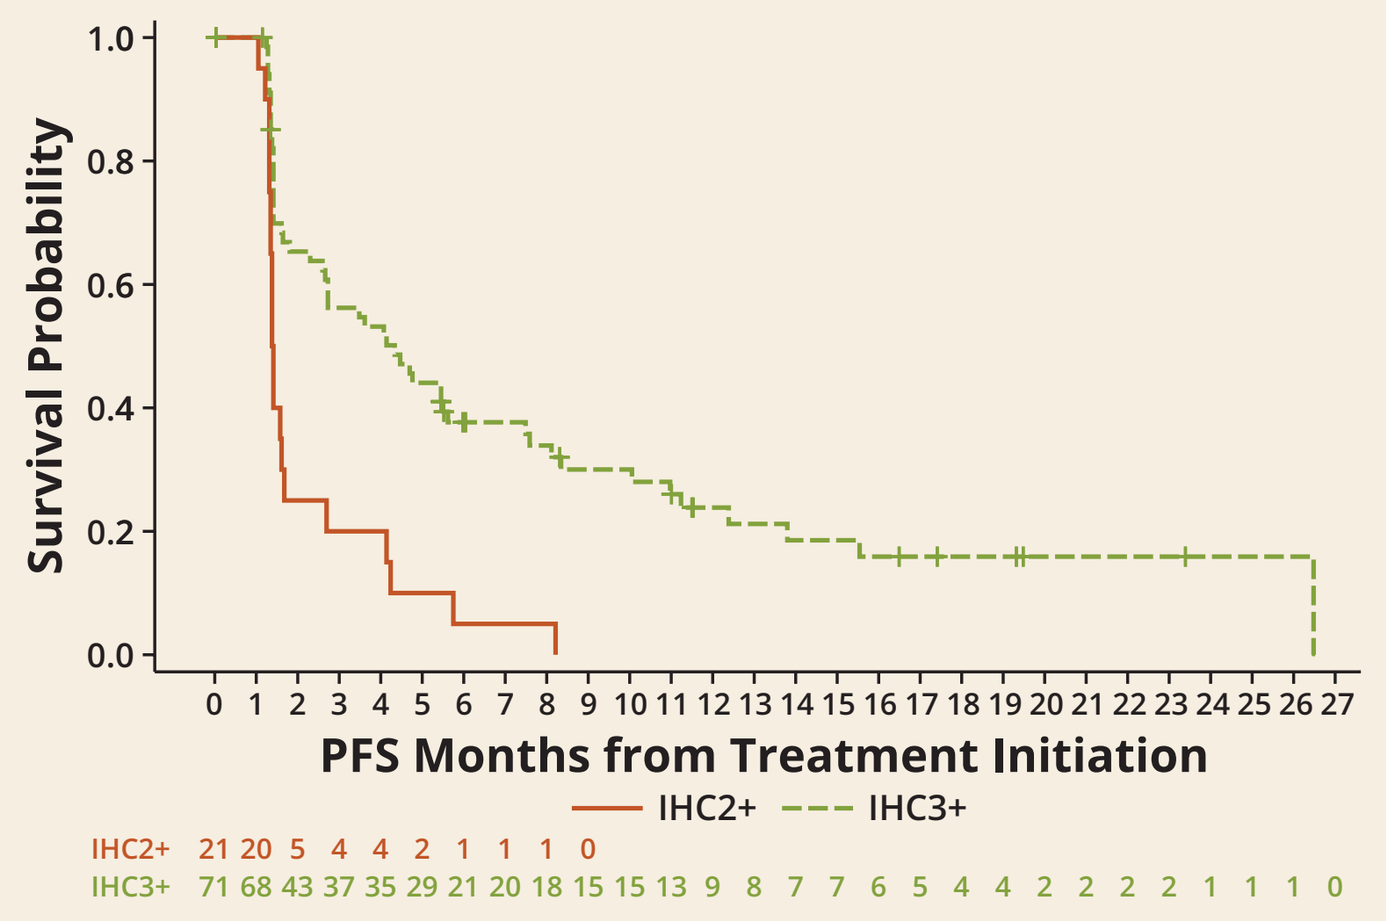
Survival Probability
PFS Months from Treatment Initiation
IHC2+
IHC3+
IHC2+ 21 20 5 4 4 2 1 1 1 0
IHC3+ 71 68 43 37 35 29 21 20 18 15 15 13 9 8 7 7 6 5 4 4 2 2 2 1 1 1 0

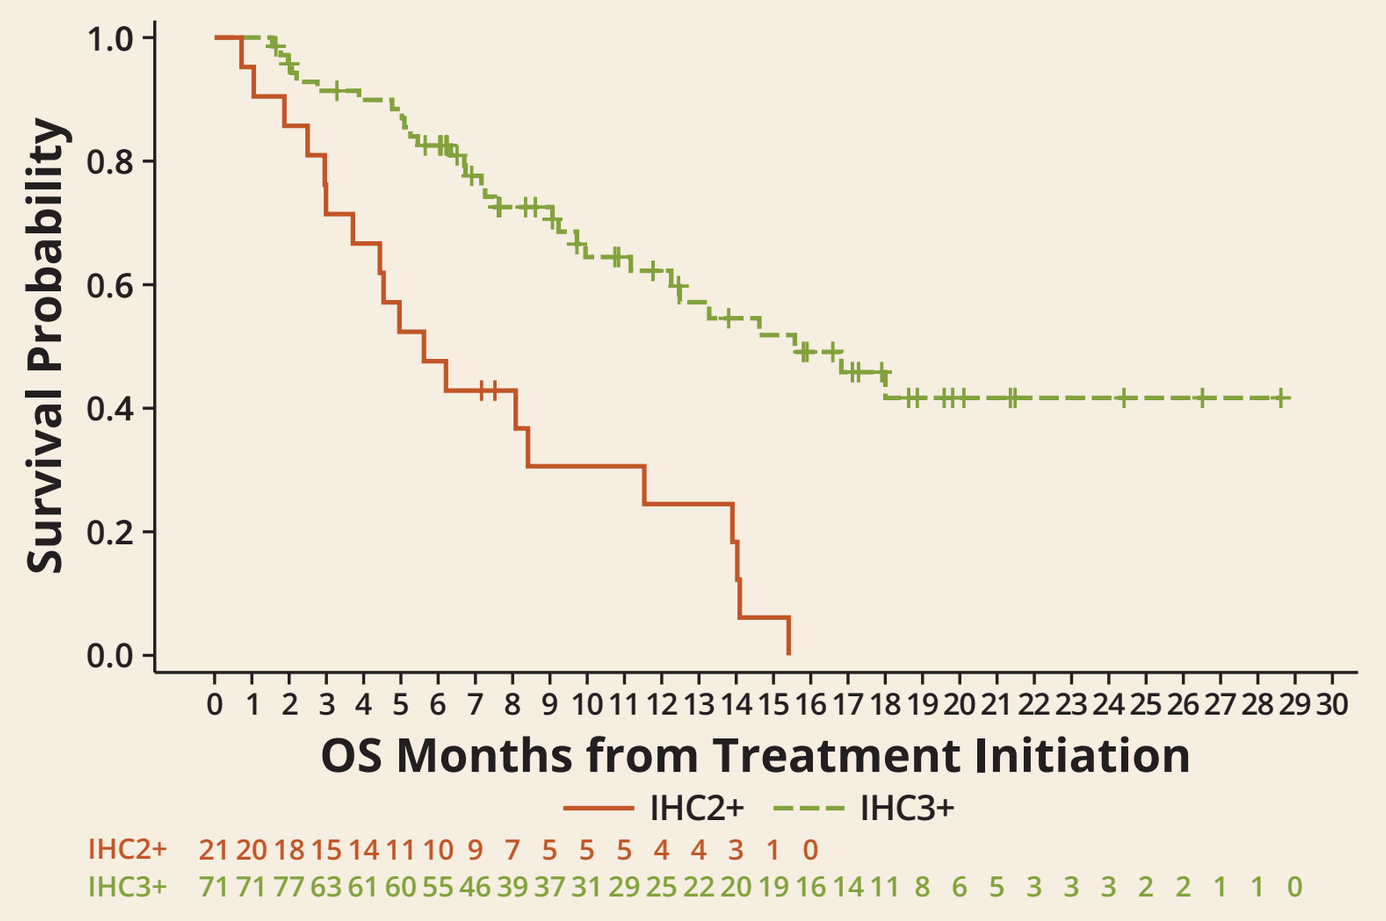
Survival Probability
OS Months from Treatment Initiation
IHC2+
IHC3+

ERBB2/HER2 Expression is Associated with HER2 3+ IHC

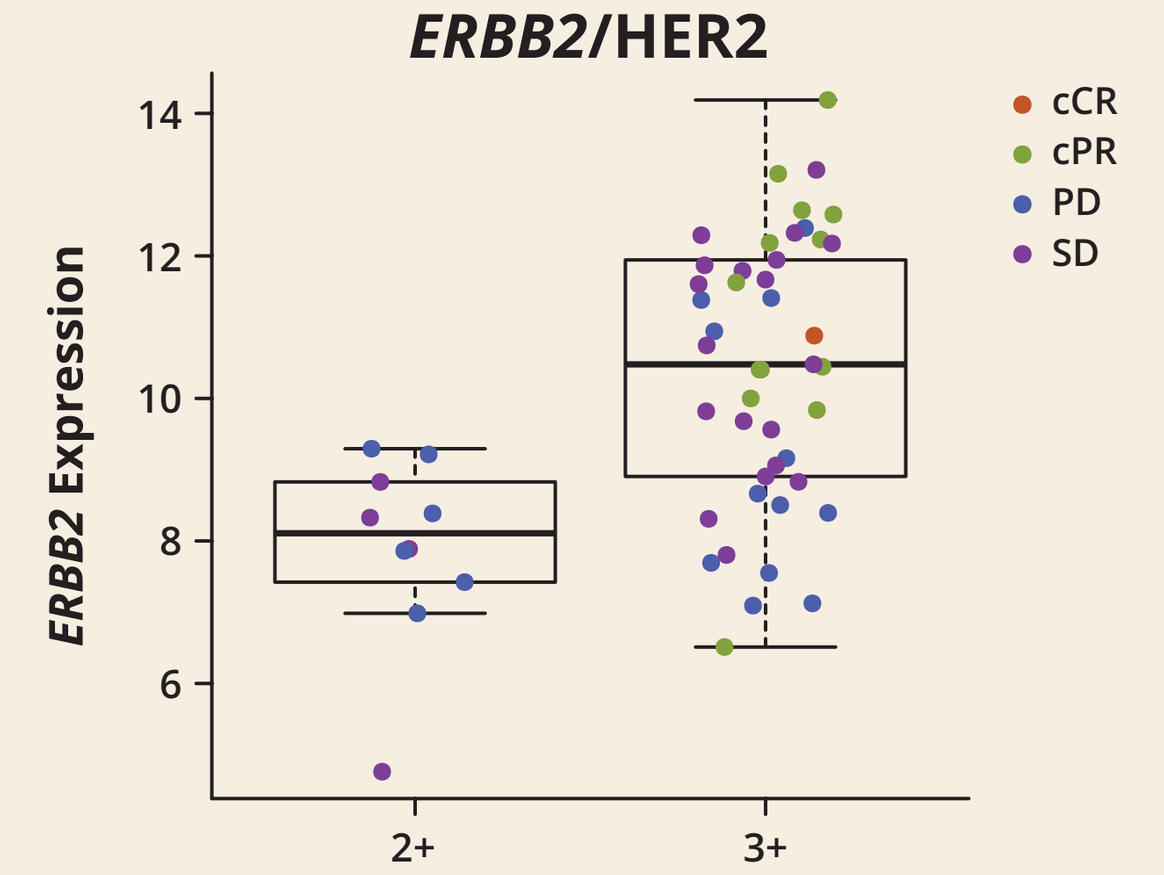
ERBB2/HER2
ERBB2 Expression
cCR
cPR
PD
SD
2+
3+

- *ERBB2* mRNA expression was 5.6 times higher in patients with HER2 IHC3+ compared with IHC2+ tumors

PD-L1 Positive IHC Is Associated with Interferon-related Signatures

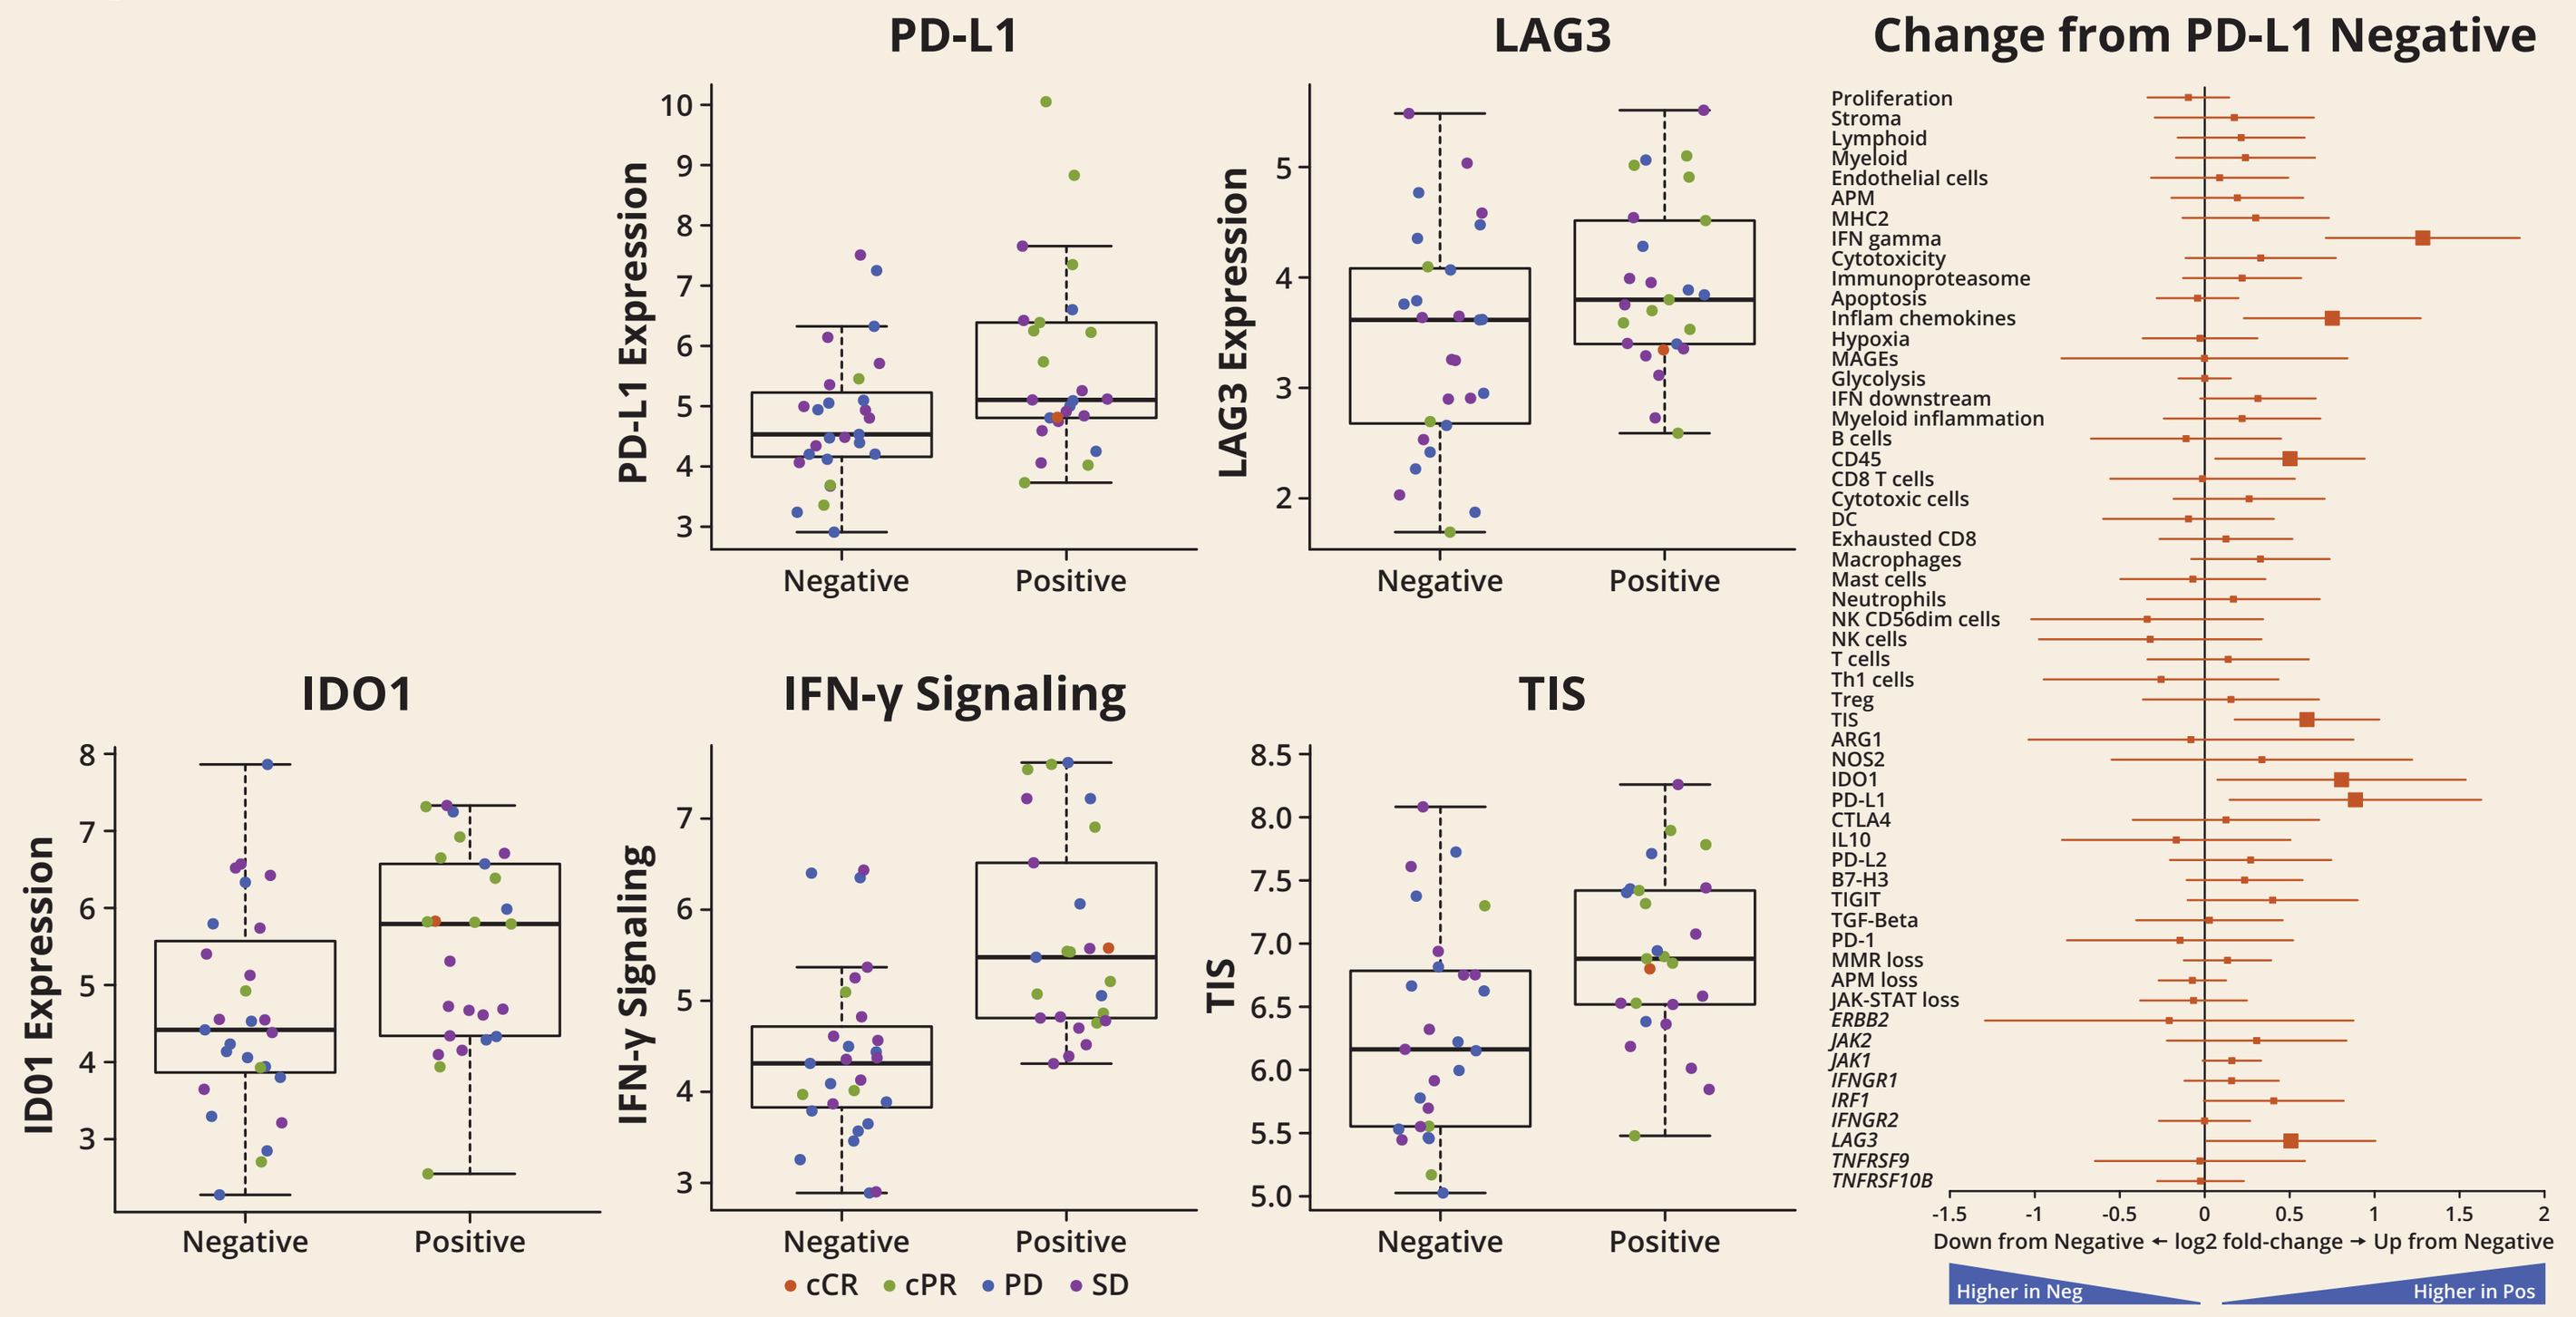
PD-L1
PD-L1 Expression
LAG3
LAG3 Expression
Change from PD-L1 Negative
IDO1
IDO1 Expression
IFN-γ Signaling
TIS
Negative
Positive
cCR
cPR
PD
SD
Down from Negative ← log2 fold-change → Up from Negative
Higher in Neg
Higher in Pos

- PD-L1-positive tumors had higher expression of PD-L1, IFN-γ signaling, LAG3, IDO1, inflammatory chemokine and tumor inflammation signature (TIS) scores

ERBB2/HER2 Expression is Associated with Gastric Tumors

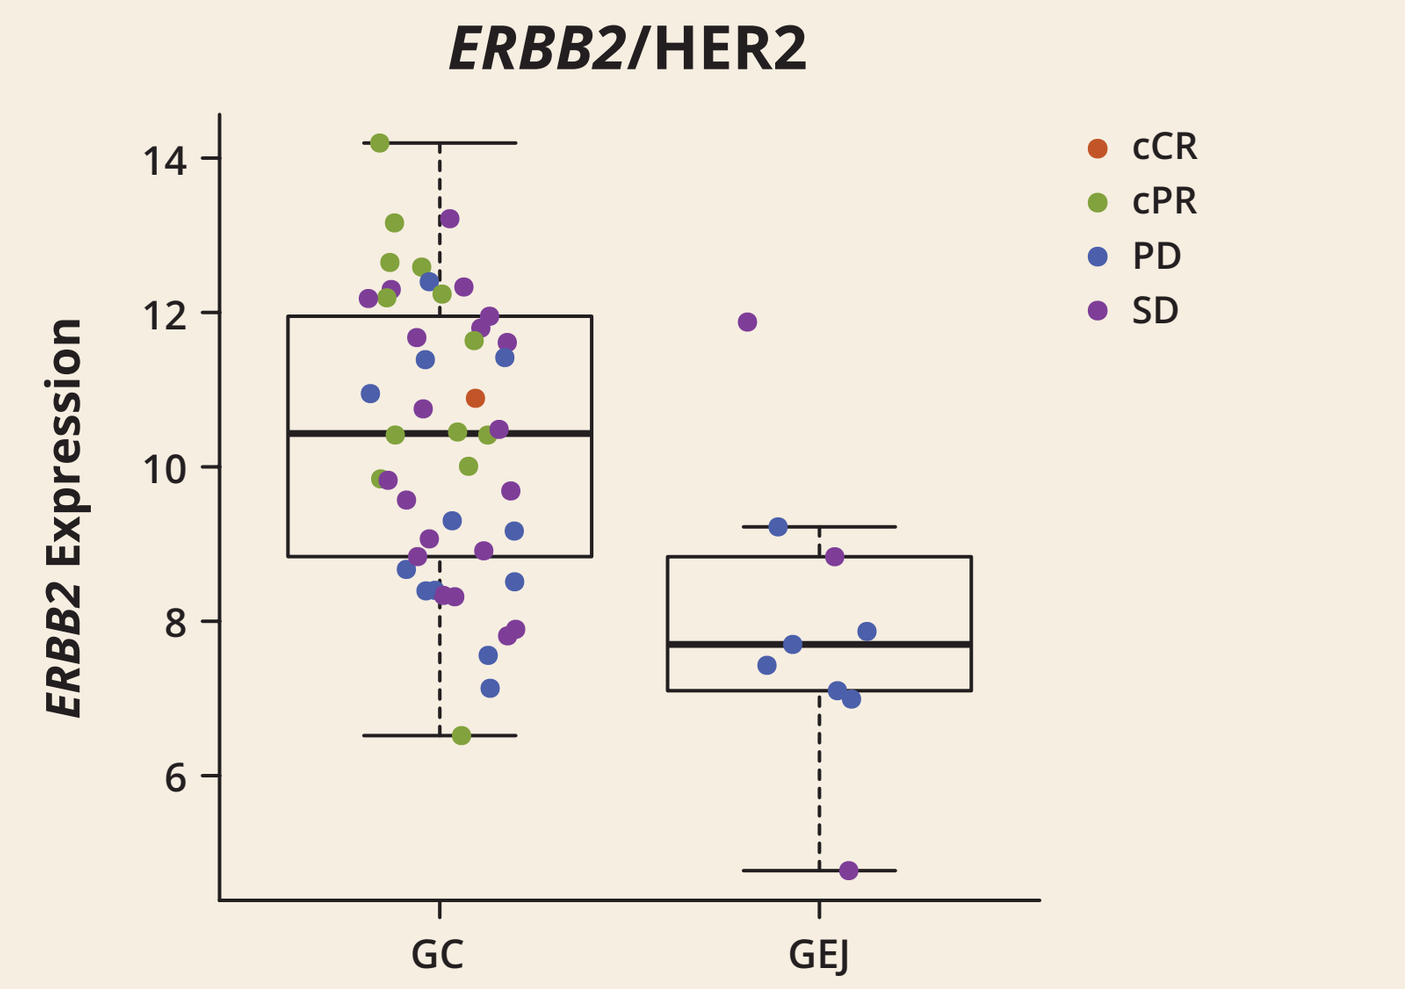
ERBB2/HER2
ERBB2 Expression
cCR
cPR
PD
SD
GC
GEJ

- Gastric tumors xpressed higher levels of *ERBB2* (5.25 FC, p<0.001) compared with GEJ tumors

Differential Gene Expression in Gastric vs. GEJ Tumors

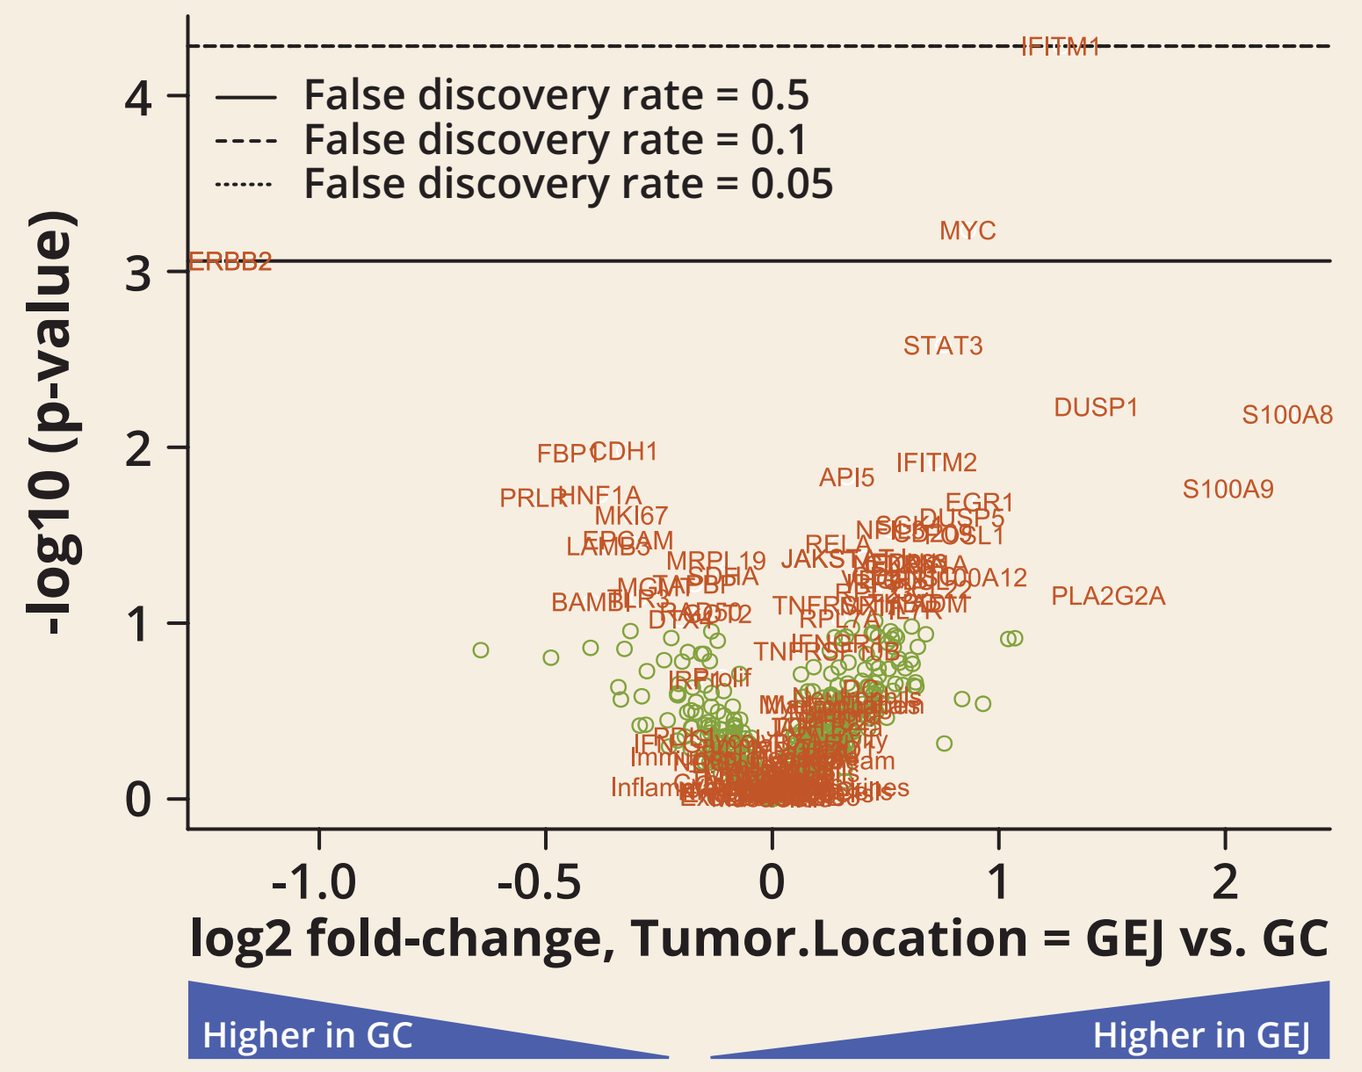
False discovery rate = 0.5
False discovery rate = 0.1
False discovery rate = 0.05
-log10 (p-value)
log2 fold-change, Tumor.Location = GEJ vs. GC
Higher in GC
Higher in GEJ

Gene/Signature	Fold Change	p-value
IFITM1	2.420	0.0001
MYC	1.820	0.0006
ERBB2	5.252	0.0009
STAT3	1.687	0.0026
DUSP1	2.699	0.0059
S100A8	4.841	0.0065
CDH1	1.574	0.0105
FBP1	1.863	0.0108
IFITM2	1.653	0.0121
API5	1.257	0.0148
S100A9	4.036	0.0172
HNF1A	1.695	0.0187
PRLR	2.073	0.0193
EGR1	1.888	0.0205
MKI67	1.538	0.0244
DUSP5	1.787	0.0251
SGK1	1.522	0.0277
NFIL3	1.435	0.0295
CD209	1.632	0.0307
FOSL1	1.804	0.0316
EPCAM	1.554	0.0337
RELA	1.224	0.0355
LAMB3	1.651	0.0365
MRPL19	1.187	0.0441
EDN1	1.501	0.0452
NFKBIA	1.473	0.0462
CDKN1A	1.554	0.0464

- GEJ tumors had lower expression of *ERBB2* and higher expression of *IFITM1*, *MYC* and *STAT3*, and less clinical responses

Discussion

- We show for the first time that GEA with high *ERBB2* expression have an inflamed TME
- Our initial data describe potential immunologic differences between GC and GEJ tumors

Reference

1.

This study was sponsored by MacroGenics, Inc. Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

Presented at the 2019 Annual Congress of the European Society for Medical Oncology, September 27–October 1, 2019, Barcelona, Spain

sergio.rutella@ntu.ac.uk